Possible Factors Leading to Inappropriate Discontinuation of Kuvan® (sapropterin dihydrochloride) with Minimum 3 Months of Continued Therapy

Michael Imperiale, Ruth Suter, Elaina Jurecki, Chris Schelling, Sean Turbeville

BioMarin Pharmaceutical Inc., Novato, CA, USA

OBJECTIVES/PURPOSE

A small percentage of patients who have received Kuvan (sapropterin dihydrochloride) for the management of their phenylketonuria (PKU) are occasionally observed to discontinue treatment after clinical effectiveness has been established. A phone survey was conducted to identify why these patients discontinue their prescribed Kuvan therapy after having been on treatment for a minimum of 3 months.

METHODS

Patients were identified during a review of the BioMarin Patient and Physician Services (BPPS) database. Patients that were receiving Kuvan therapy for at least 3 months, but whose last 30-day supply of medication had not been refilled, were considered to have discontinued therapy and were included in the study. Patients who were 18 years of age or older were contacted directly by telephone and invited to participate in a brief survey. If the patient was younger than 18 years, an adult family member was invited to participate. Survey questions were designed to gather information about the duration of therapy, reasons for discontinuation, insurance coverage, length of treatment, efficacy, safety and tolerance, convenience of administration, cost, and willingness to reinitiate therapy.

RESULTS

108 patients who met the inclusion criteria were identified from the BPPS database. Of these patients, 36 (33% of total) received new drug supply shortly after 30 days and were excluded from the interview process. Six patients were thought to have a gap in refill, though this was due to changing over from a 30-day to a 90-day supply of the medication. Figure 1 shows the PKU interview summary for the patients who met the inclusion criteria.

Among the remaining 66 patients (61% of total), 18 participated in the survey for an overall response rate of 27% (18/66). The median patient age was 16.5 years, ranging from 3 to 47 years. There were no significant differences in demographics between survey participants and non-participants, and participants were equally distributed by geography. Five patients (28%) discontinued treatment due to non-response related to “perceived lack of efficacy” and/or “increasing phe levels”. (One of these patients discontinued therapy despite a reduction in blood Phe levels from 16 mg/dL to 9 mg/dL. In two of these patients, premature liberalization of the patient’s Phe-restricted diet may have confounded Kuvan response). Other reasons for discontinuation included “lack of perceived benefit” (28%) mostly due to the inconvenience of daily drug administration and noncompliance (11%) attributed to overall “pill burden.” One child experienced a significant reduction in blood Phe level (12 mg/dL to 1 mg/dL) with an adverse event (lack of concentration) that required temporary discontinuation of treatment. Figure 2 illustrates the reasons for discontinuation of Kuvan.

15 of the 18 participants (83%) indicated a willingness to restart Kuvan once their specific issues had been addressed (Figure 3). Of the remaining 3 who were not willing to restart Kuvan, 2 (11%) were from the group classified as non-responders and 1 (6%) was from the group that discontinued due to perceived lack of treatment benefit.

CONCLUSION

The majority of patients discontinuing therapy expressed interest in restarting the medication despite concerns. Appropriate case management and continued outreach to improve education regarding the net clinical benefit of chronic therapy and awareness of patient assistance programs such as BioMarin’s Kuvan Patient Assistance Program and the National Organization for Rare Disorders (NORD) PKU Premium/Co-Payment Assistance Program may prevent unnecessary early discontinuation from therapy. BioMarin offers patient-oriented education materials to clinicians interested in customizing patient education about PKU and Kuvan therapy. BPPS and NORD offer support services to individuals who require assistance with treatment access, insurance coverage, and reimbursement-related issues. These patient education and assistance programs may be especially beneficial to the patients who cited reasons for discontinuation due to “insurance issues,” all of whom indicated that they would consider trying Kuvan again as long as insurance can cover costs. Over time it will be important to understand the factors associated with long-term adherence and to adjust educational, individual case management and other intervention strategies accordingly.

SURVEY PARTICIPANTS BY STATE

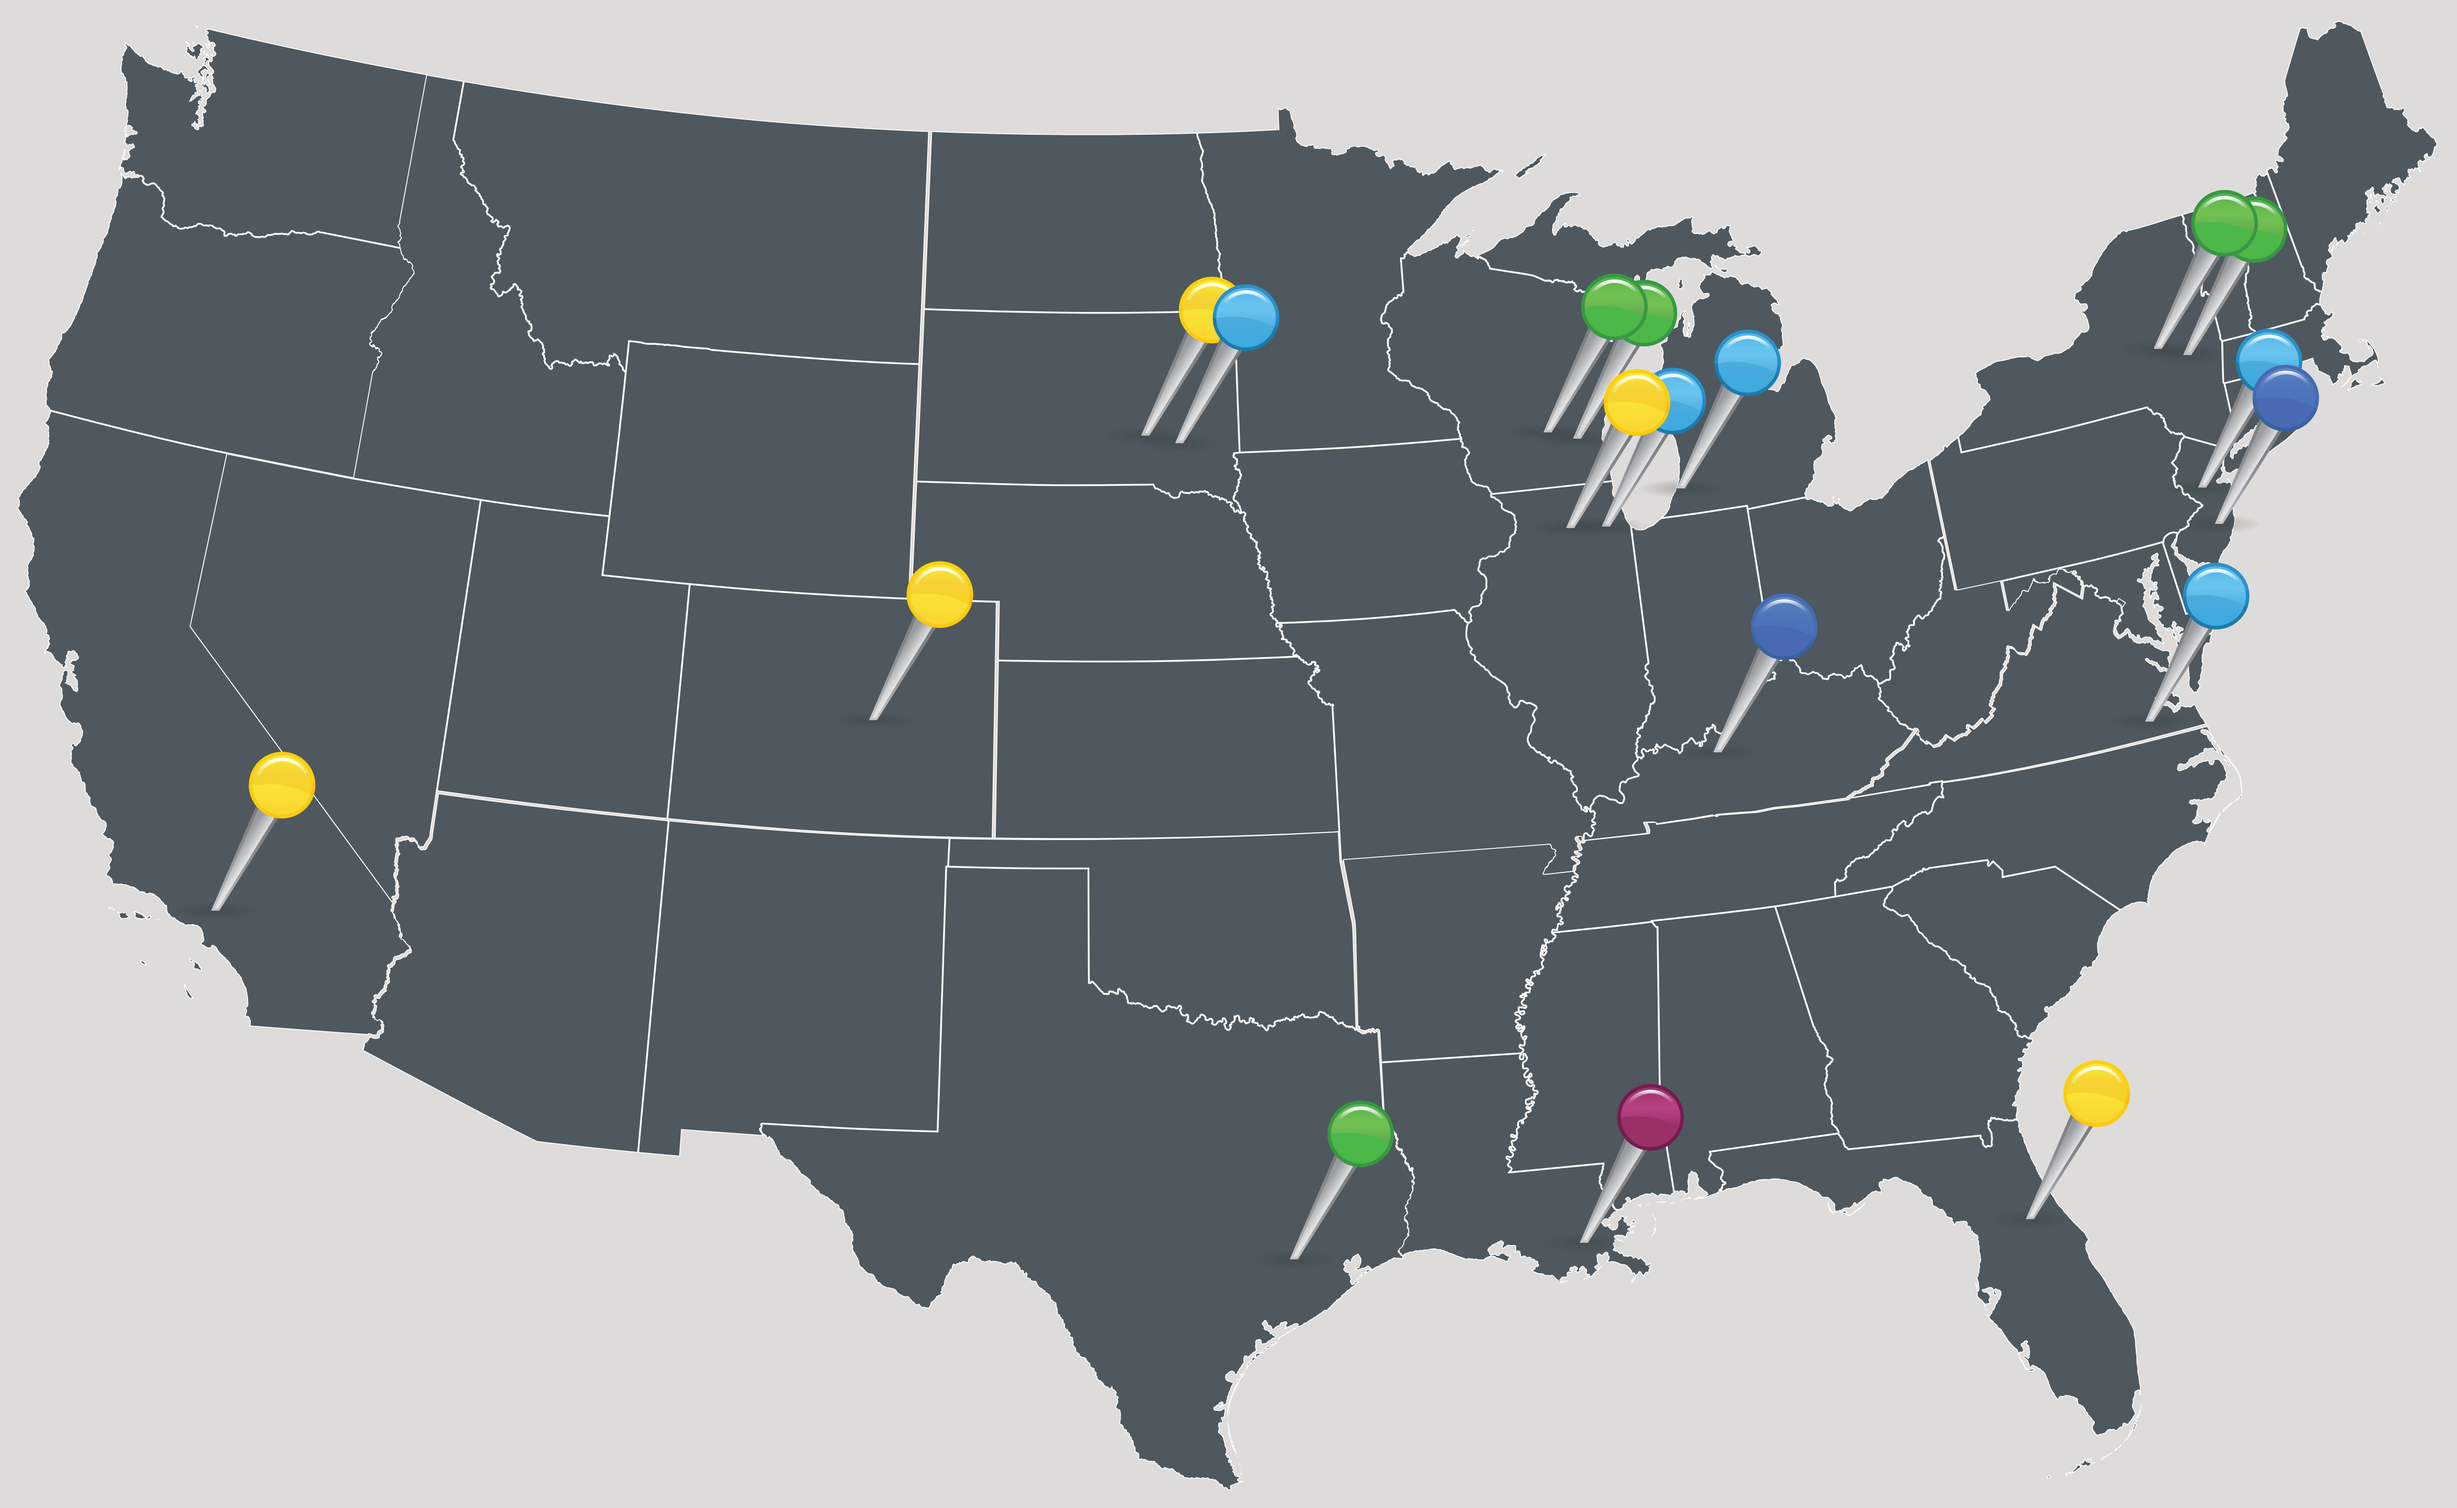

1	Gainesville, FL	1	Denver, CO	1	Norfolk, VA	1	Houston, TX
1	Chicago, IL	1	Detroit, MI	1	Chicago, IL	1	Louisville, KY
1	Sioux Falls, SD	1	Newark, NJ	2	Albany, NY	1	Camden, NJ
1	Los Angeles, CA	1	Sioux Falls, SD	2	Milwaukee, WI	1	New Orleans, LA

Reasons for discontinuation

Insurance Issues | Non-responders | No perceived benefit of therapy | Non-compliant patients | Experienced side effects

ACKNOWLEDGEMENTS

The authors acknowledge Kathy Tam, BScPhm, of MediResource Inc., who provided medical writing assistance in drafting the poster, Joanne Lee, BSc, of MediResource Inc., who provided editorial assistance, and Catherine Smith of MediResource Inc., who prepared the graphical layout of the poster. BioMarin Pharmaceutical Inc. funded the study and the medical writing, editorial, and graphics assistance for the poster.

FIGURE 1: PKU INTERVIEW SUMMARY
% of discontinued patients who met inclusion criteria (N=108)

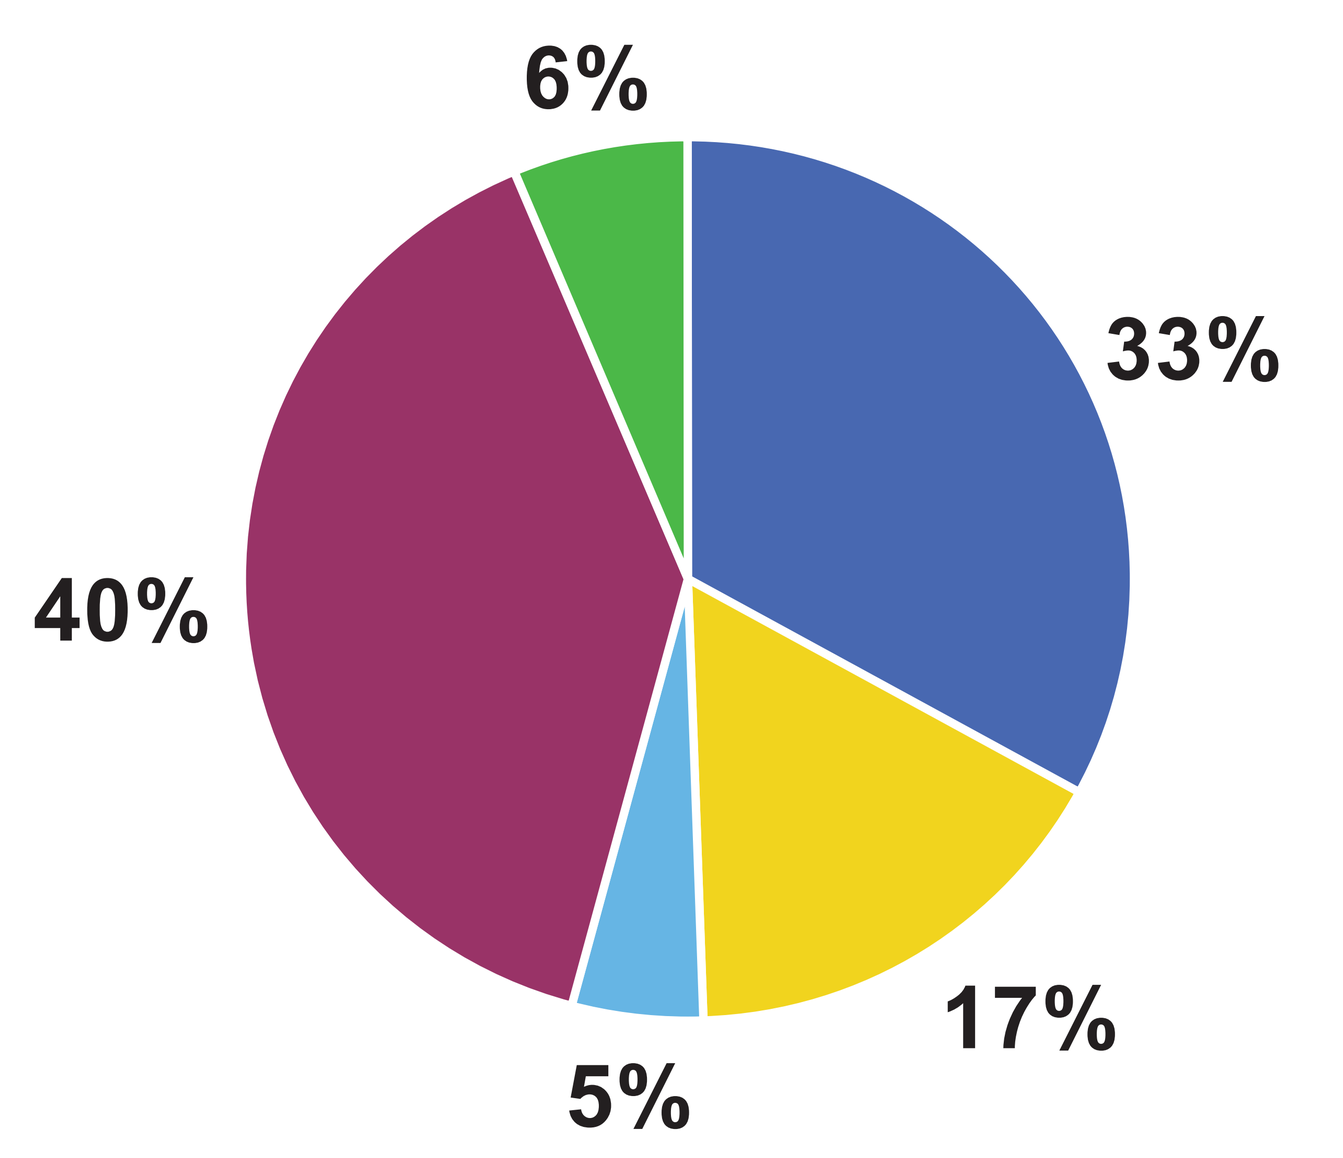
6%
33%
40%
17%
5%

- Interviewed—gap in refill related to pts receiving 90-day vs. 30-day supply
- Not interviewed—received new supply
- Interviewed
- Not interested
- Did not respond to survey

FIGURE 2: REASONS FOR DISCONTINUATION
% of Patients who participated in survey (N=18)

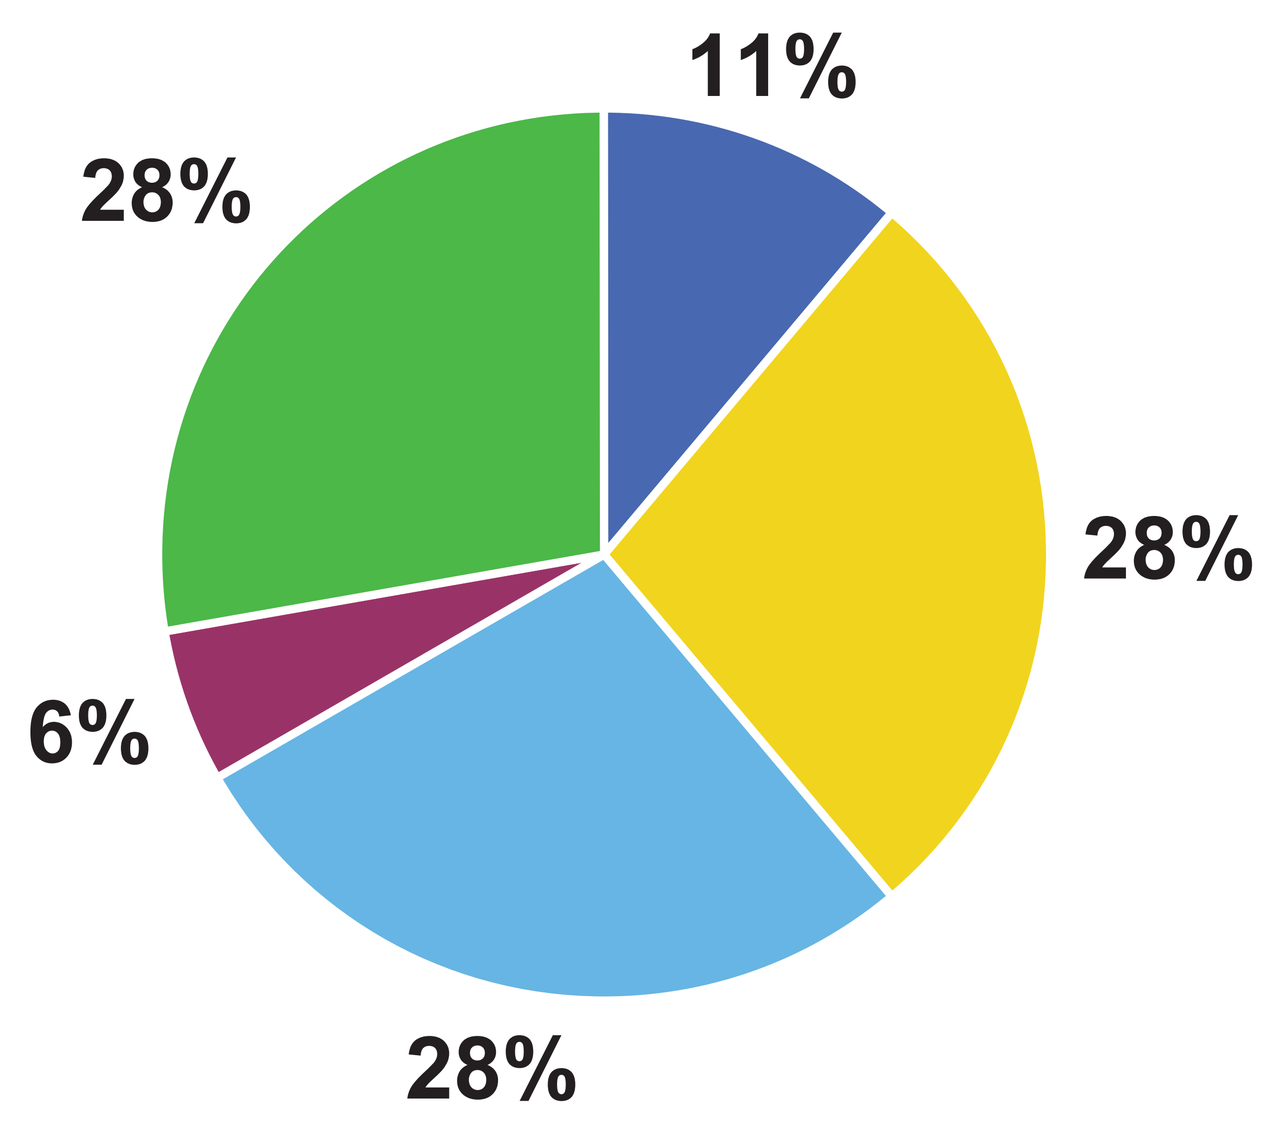
11%
28%
28%
6%
28%

- No perceived benefit of Tx
- Non-compliant patients
- Insurance issues
- Non-responders
- Experienced side effects

*Percentages add up to >100% due to rounding

FIGURE 3: WILLINGNESS TO RESTART KUVAN

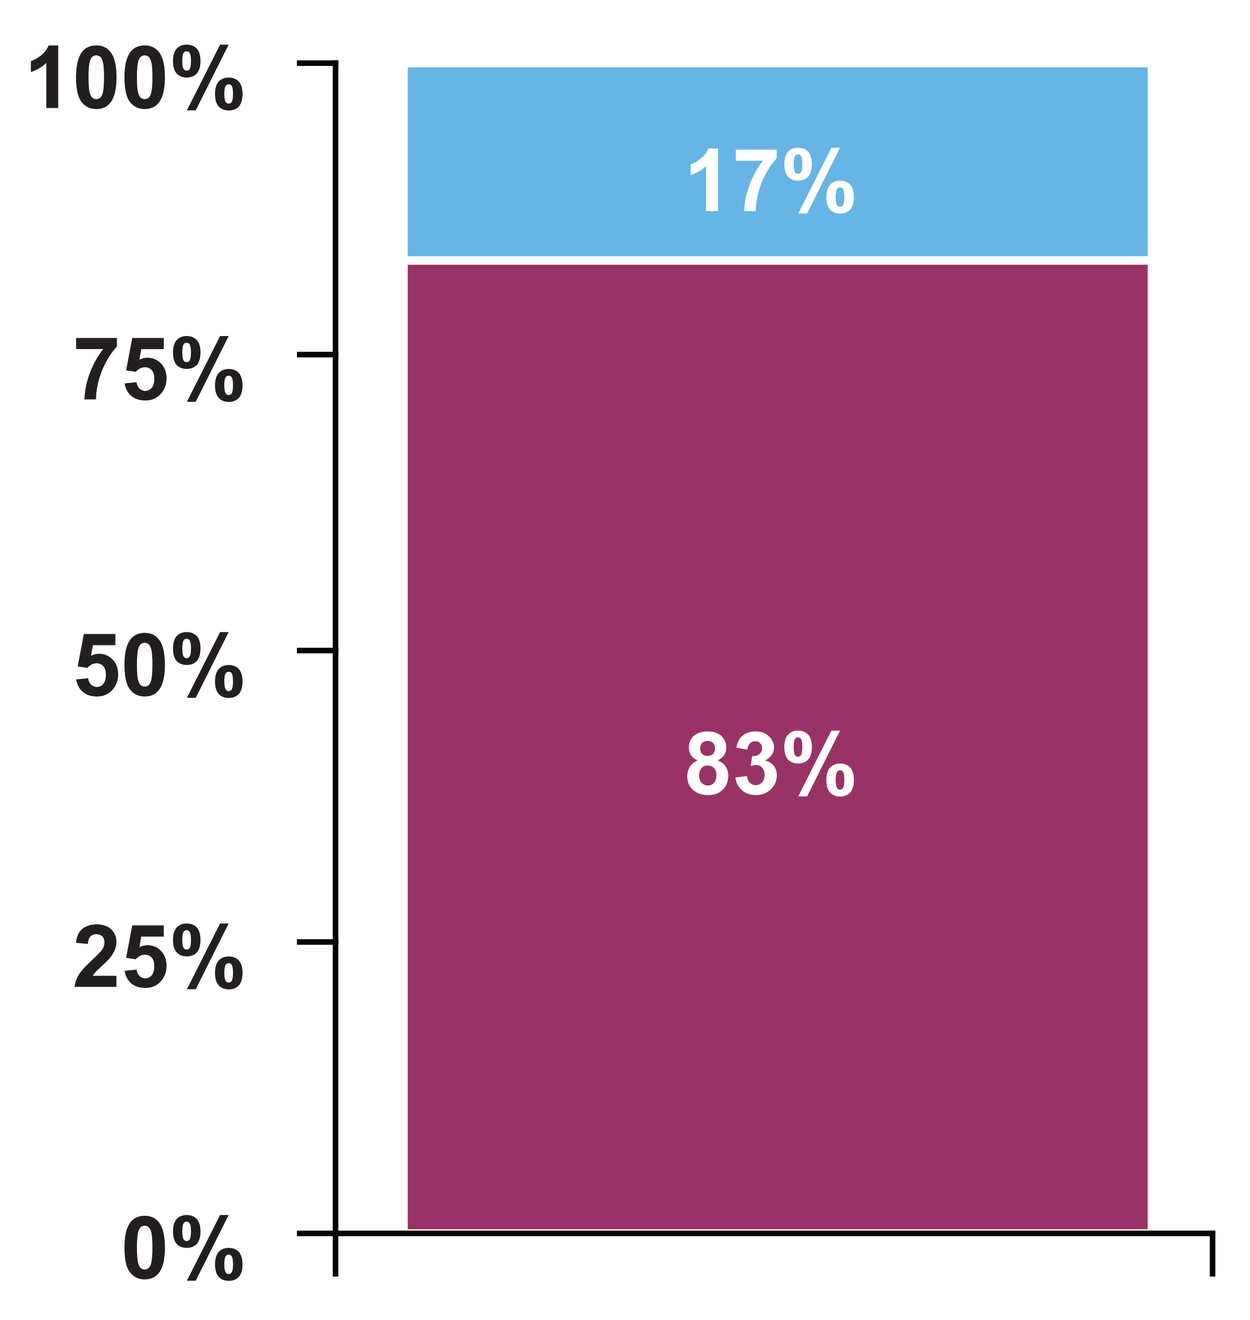
100%
75%
50%
25%
0%
17%
83%

- Would NOT restart Kuvan® Tx
- Would restart Kuvan® Tx